axoltis pharma
www.axoltis.com

SCO-spondin derived peptide, innovative drug candidates for the treatment of Alzheimer's disease

Juliette Le Douce[1], Nathalie Delétage[1], Sighild Lemarchant[1], Yann Godfrin[1,2]
[1]Axoltis Pharma, Lyon, France, [2]GODFRIN Life-Sciences, Caluire-et-Cuire, France

P193
Contact: jledouce@axoltis.com

INTRODUCTION

Alzheimer's disease (AD) is a neurodegenerative disease affecting million people who suffer from the progressive deterioration of cognitive functions including learning and memory. The few approved treatments such as donepezil (DPZ) are limited to the symptomatic control of AD, therefore there is an urgent need to develop a disease-modifying treatment to halt AD-induced cognitive deficits. The subcommissural organ (SCO)-spondin is a brain-specific glycoprotein which contributes to neuronal development. It was shown that a SCO-spondin-derived peptide named NX has neuroprotective effects (Delétage, Le Douce *et al*, 2021). Here, we sought to evaluate the protective effects of NX on learning and memory in a mouse model of AD.
NX peptide was tested as a single treatment, a combinatory treatment or a second-line treatment following DPZ loss of efficacy in a mouse model characterized by cognitive alterations.

METHODS

Mouse model of AD. The amyloid-beta [25–35] ($A\beta_{25\text{-}35}$) and control scrambled $A\beta_{25\text{-}35}$ (Sc.Aβ) were injected in the right lateral ventricle in male Swiss mice on day 1 (9 nmol per mouse). **Treatments.** NX was administered at 0.1, 1, 2, 3.75 or 30 mg/kg. Donepezil (DPZ) was administered orally at 1 mg/kg (positive control). **Behavioral tests.** Memory was evaluated using three tests: the Y-maze (YM) test to assess short-term working memory, the passive avoidance test (STPA) to assess contextual long-term memory and the Morris water maze (MWM) to assess spatial learning and recall memory. **Biochemical analysis.** Prefrontal cortices and hippocampi from mouse brains were dissected and analyzed for different AD biomarkers.

RESULTS

NX peptide prevents memory loss as a monotherapy and as a combinatory treatment with DPZ.

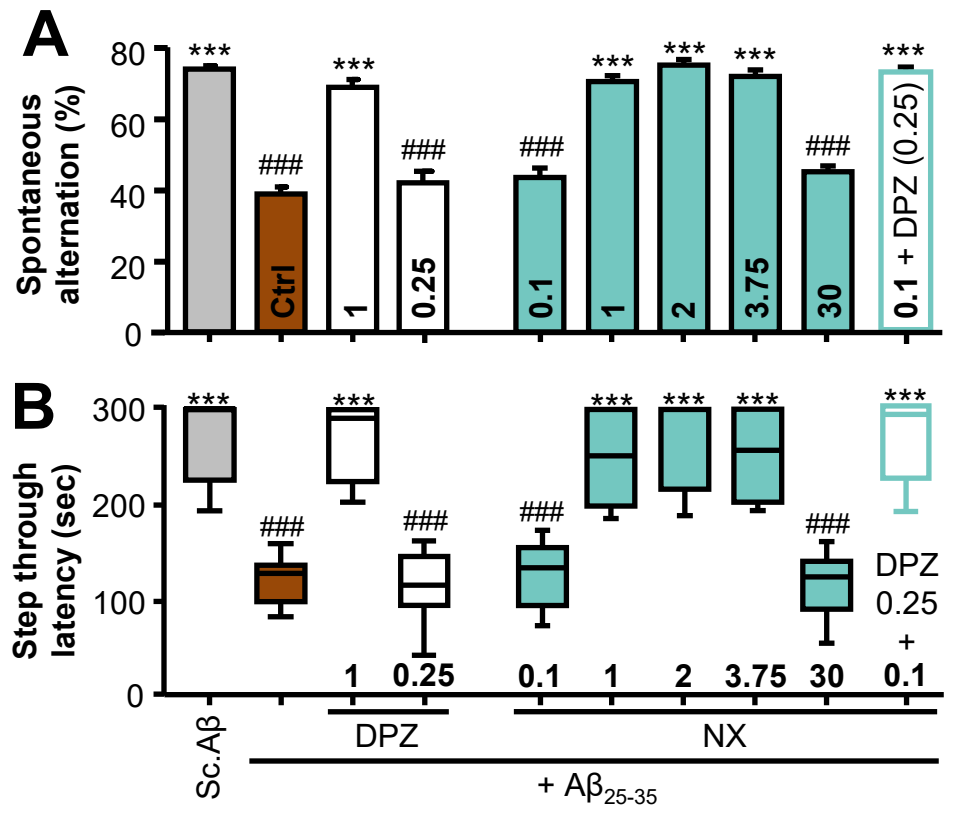
A
Spontaneous alternation (%)
Ctrl
1
0.25
0.1
1
2
3.75
30
0.1 + DPZ (0.25)
B
Step through latency (sec)
DPZ
0.25
+
0.1
Sc.Aβ
DPZ
NX
+ Aβ25-35

Vehicle, DPZ or NX was administered from day 1 to day 10 and doses are expressed in mg/kg. (**A**) YM was performed on D8 and (**B**) STPA on day 9-10. One-way ANOVA followed by Dunnett's multiple comparison test for (**A**). Kruskal-Wallis followed by Dunn's multiple comparison test for (**B**). ***$p<0.001$ vs. $A\beta_{25\text{-}35}$ / vhc group, ###$p<0.001$ vs. Sc.Aβ / vhc group. n=11-18 mice/group.

NX peptide prevents spatial learning impairment and alteration of recall memory as a monotherapy.

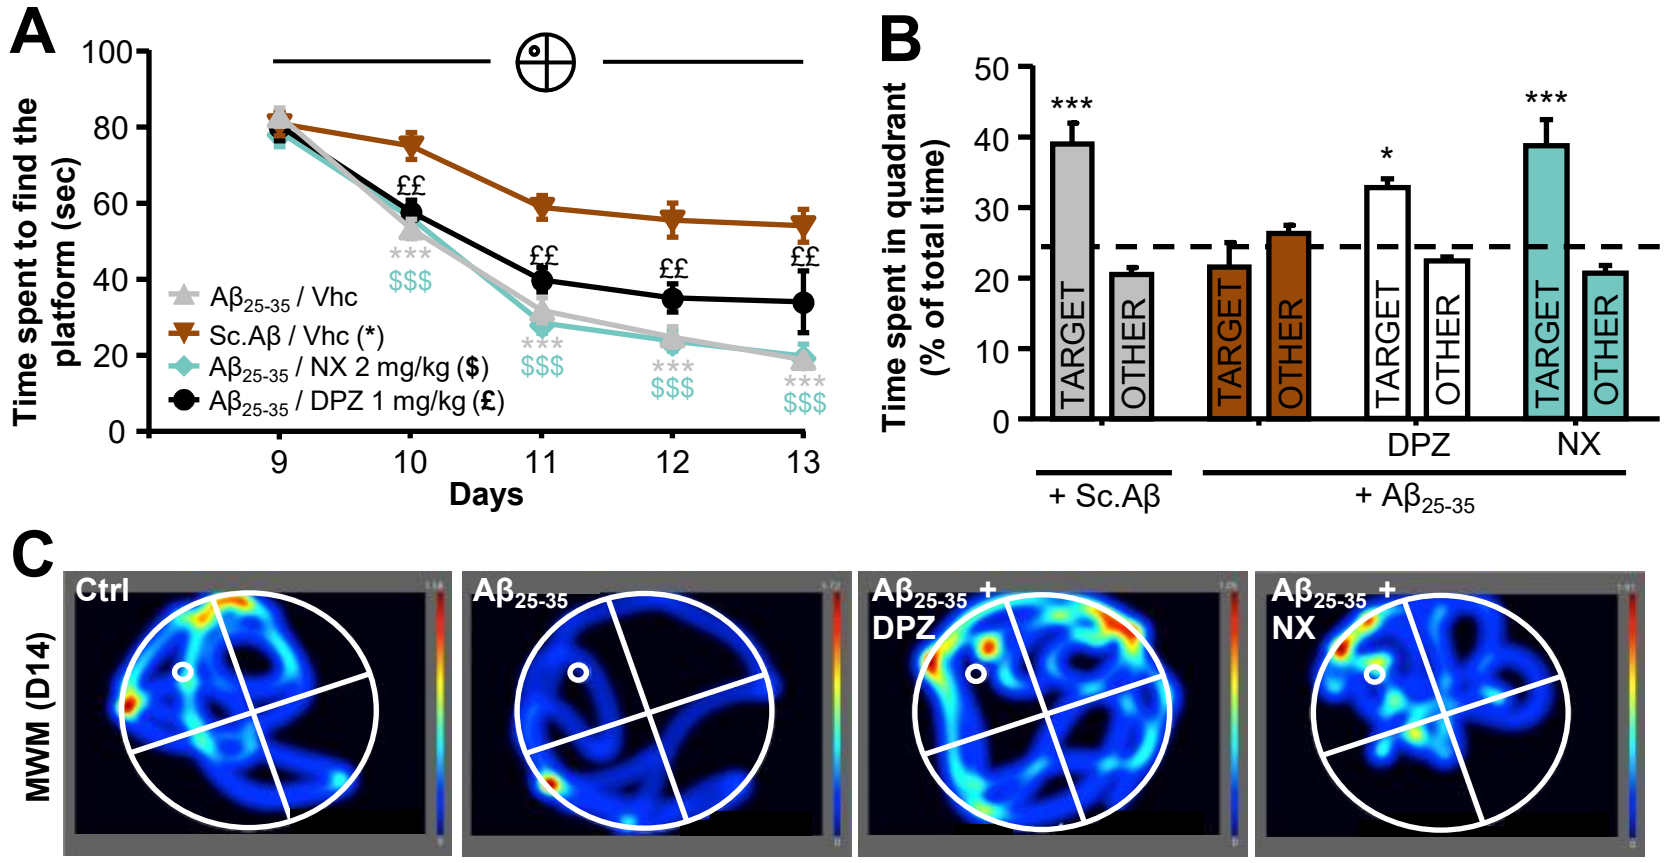
A
Time spent to find the platform (sec)
Days
Aβ25-35 / Vhc
Sc.Aβ / Vhc (*)
Aβ25-35 / NX 2 mg/kg ($)
Aβ25-35 / DPZ 1 mg/kg (£)
B
Time spent in quadrant (% of total time)
TARGET
OTHER
DPZ
NX
+ Sc.Aβ
+ Aβ25-35
C
MWM (D14)
Ctrl
Aβ25-35
Aβ25-35 + DPZ
Aβ25-35 + NX

Vehicle, DPZ or NX was administered from day 1 to day 14. MWM was performed from D9 to D13 for spatial learning and the probe test was performed on D14. Two-way ANOVA followed by Tukey multiple comparison test for (**A,B**). *$p<0.05$, **$p<0.01$, ***$p<0.001$ vs. $A\beta_{25\text{-}35}$ / vhc group. n=8-12 mice/group.

NX peptide completely restores AD biomarkers.

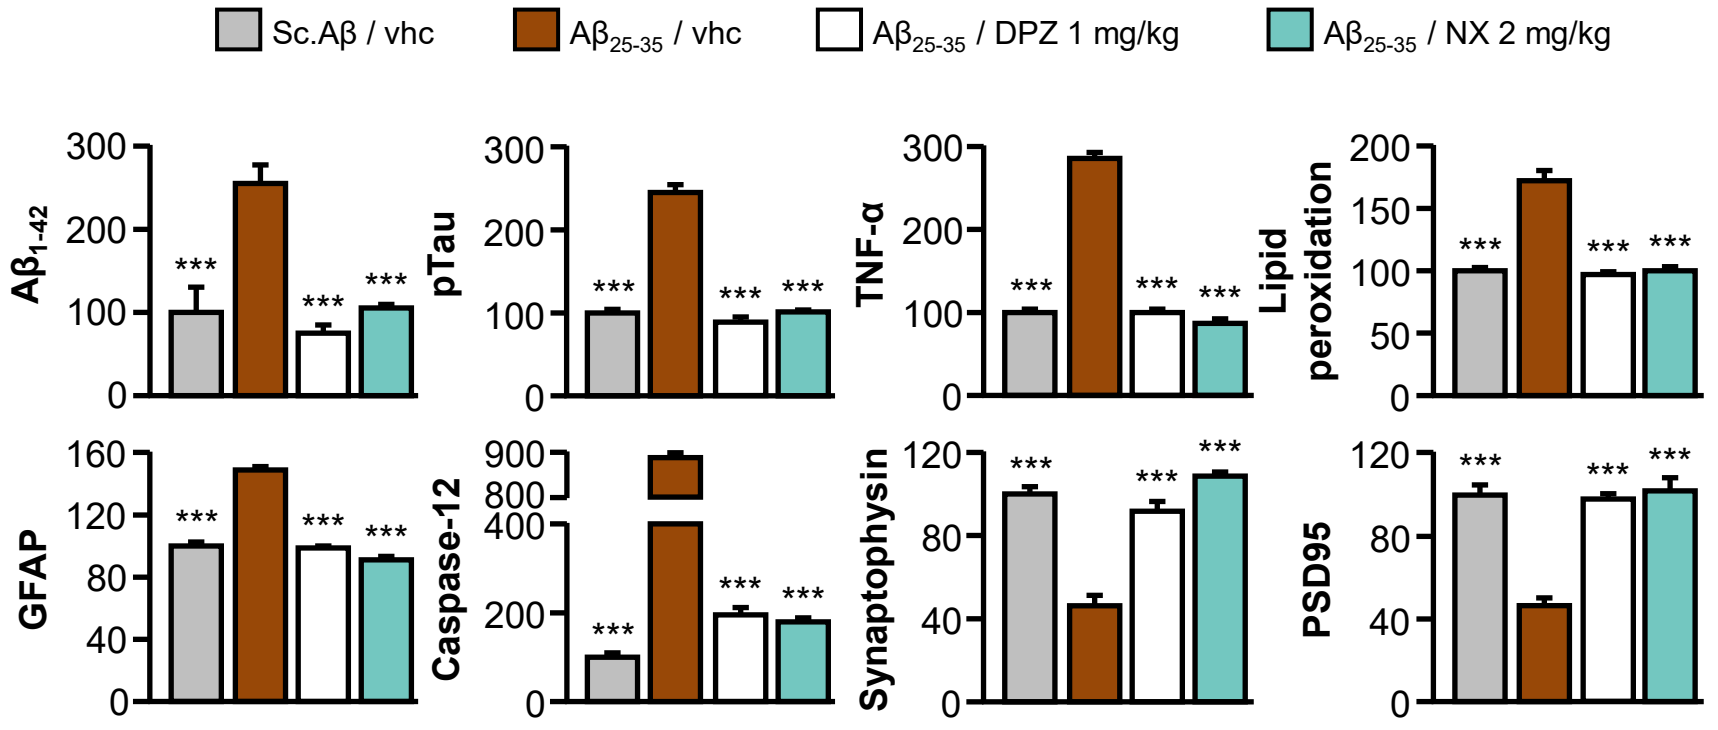
Sc.Aβ / vhc
Aβ25-35 / vhc
Aβ25-35 / DPZ 1 mg/kg
Aβ25-35 / NX 2 mg/kg
Aβ1-42
pTau
TNF-α
Lipid peroxidation
GFAP
Caspase-12
Synaptophysin
PSD95

Vehicle, DPZ or NX was administered from day 1 to day 11 and brain sampled on day 11. Data are expressed as percent of control. One-way ANOVA followed by Dunnett's multiple comparison test for all graphics. ***$p<0.001$ vs. $A\beta_{25\text{-}35}$ / vhc group. n=5-6 mice/group.

NX peptide prevents memory loss without loss of efficacy and a 4-week treatment restores and maintains cognitive function up to 4 months.

NX peptide, as a synergic treatment with DPZ, has a sustained activity on memory loss over 120 days.

NX peptide is an efficient second-line treatment following DPZ loss of efficacy.

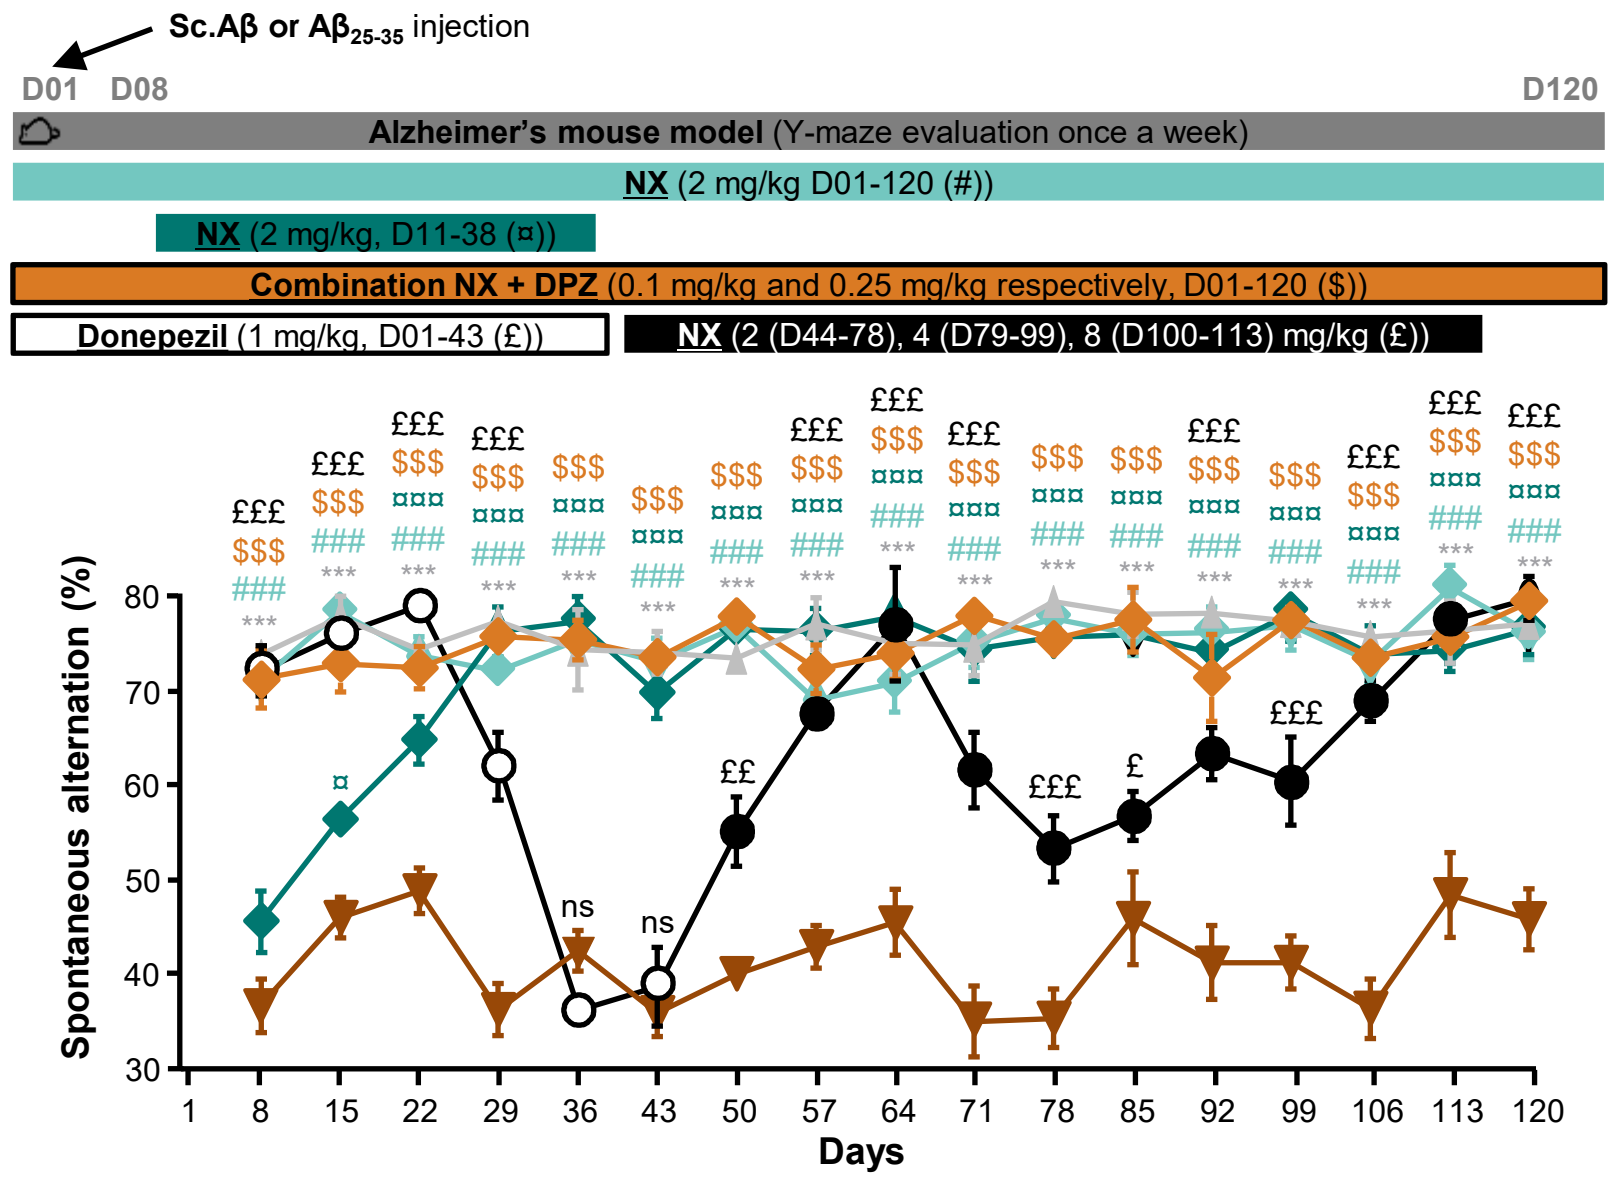
Sc.Aβ or Aβ25-35 injection
D01
D08
D120
Alzheimer's mouse model (Y-maze evaluation once a week)
NX (2 mg/kg D01-120 (#))
NX (2 mg/kg, D11-38 (¤))
Combination NX + DPZ (0.1 mg/kg and 0.25 mg/kg respectively, D01-120 ($))
Donepezil (1 mg/kg, D01-43 (£))
NX (2 (D44-78), 4 (D79-99), 8 (D100-113) mg/kg (£))
Spontaneous alternation (%)
Days

Two-way ANOVA followed by Sidak's multiple comparison test. ¤$p<0.05$, ***, ###, $$$, £££, ¤¤¤$p<0.001$ vs. $A\beta_{25\text{-}35}$ / vhc group. n=5-6 mice/group.

CONCLUSION

NX peptide is a promising disease-modifying drug candidate showing a strong and sustained efficacy as a monotherapy, in combination with DPZ or as a second-line treatment following DPZ loss of efficacy on an *in vivo* model of AD by acting on amyloid, tau, neuroinflammation, oxidative stress, and memory loss.

Please check out our other posters about NX efficacy in excitotoxicity (P625) and amyotrophic lateral sclerosis (P594)!

Acknowledgments: Thanks to the Ministère de l'Economie et des Finances and Région Auvergne-Rhône-Alpes for their financial support [DOS0082366 and DOS0082367] and to Amylgen for experiments. **Disclosure:** YG is chief executive officer and a shareholder of Axoltis Pharma – JLD, ND and SL are employees of Axoltis Pharma. **Publication:** Delétage, Le Douce *et al.*, Neuroscience, 2021; Le Douce *et al.*, in revision